Abstract #1091

TRILACICLIB COMBINED WITH SACITUZUMAB GOVITECAN (SG) IN METASTATIC TRIPLE-NEGATIVE BREAST CANCER (mTNBC): UPDATED PHASE 2 SAFETY AND EFFICACY RESULTS

LASIKA SENEVIRATNE[1]; KATHLEEN HARNDEN[2]; SIBEL BLAU[3]; MICHAEL DANSO[4]; DAVID BERZ[5]; DELIA GUAQUETA[6]; JOHN SCHWERKOSKE[7]; JOYCE O'SHAUGHNESSY[8]; DEBRA PATT[9]; ADITYA BARDIA[10,11]; KELLY E. MCCANN[10,12]; ANDREW BEELEN[13]; JULIO RUIZ[13]; JOHN S. YI[13,*]; SARA A. HURVITZ[14]; AND MABEL MARDONES[15]

[1] LOS ANGELES CANCER NETWORK/ONEONCOLOGY, LOS ANGELES, CA, USA; [2] INOVA SCHAR CANCER INSTITUTE, FAIRFAX, VA, USA; [3] NORTHWEST MEDICAL SPECIALTIES, PUYALLUP AND TACOMA, WA, USA; [4] VIRGINIA ONCOLOGY ASSOCIATES, NORFOLK, VA, USA; [5] VALKYRIE CLINICAL TRIALS, LOS ANGELES, CA, USA; [6] MEMORIAL REGIONAL HOSPITAL, HOLLYWOOD, FL, USA; [7] MINNESOTA ONCOLOGY HEMATOLOGY P.A., WOODBURY, MN, USA; [8] TEXAS ONCOLOGY-BAYLOR CHARLES A. SAMMONS CANCER CENTER, DALLAS, TX, USA; [9] TEXAS ONCOLOGY-AUSTIN CENTRAL, AUSTIN, TX, USA; [10] DIVISION OF HEMATOLOGY/ONCOLOGY, DAVID GEFFEN SCHOOL OF MEDICINE AT THE UNIVERSITY OF CALIFORNIA LOS ANGELES, LOS ANGELES, CA, USA; [11] JONSSON COMPREHENSIVE CANCER CENTER, LOS ANGELES, CA, USA; [12] BEVERLY HILLS CANCER CARE, LOS ANGELES, CA, USA; [13] G1 THERAPEUTICS INC., RESEARCH TRIANGLE PARK, NC, USA; [14] DIVISION OF HEMATOLOGY AND ONCOLOGY, DEPARTMENT OF MEDICINE AT THE UNIVERSITY OF WASHINGTON SCHOOL OF MEDICINE/FRED HUTCHINSON CANCER CENTER, SEATTLE, WA, USA; [15] ROCKY MOUNTAIN CANCER CENTERS, DENVER, CO, USA

*AFFILIATION AT TIME OF STUDY.

INTRODUCTION

- Trilaciclib is a potent, highly selective, reversible cyclin-dependent kinase (CDK)4/6 inhibitor that enhances antitumor immunity and protects hematopoietic stem and progenitor cells (myeloprotection) when administered intravenously prior to chemotherapy[1–5]
- Results from a phase 2 study in patients with metastatic triple-negative breast cancer (mTNBC; NCT02978716) showed that administering trilaciclib prior to gemcitabine plus carboplatin provided long-term benefit by prolonging overall survival (OS) compared with administering gemcitabine plus carboplatin alone (median, 19.8 vs 12.6 months; hazard ratio, 0.37; *P* < 0.0001)[6]
 - Administering trilaciclib prior to chemotherapy also enhanced T-cell activity and modulated antitumor immunity[6]
- Sacituzumab govitecan (SG), an antibody–drug conjugate comprising a trophoblast cell surface antigen 2–directed antibody linked to the topoisomerase I inhibitor SN-38, is indicated for the treatment of adult patients with mTNBC who have received ≥ 2 prior systemic therapies, ≥ 1 of them for metastatic disease[7]
 - When internalized by tumor cells, SG induces DNA damage, leading to cell death
- In the phase 3 ASCENT trial in patients with previously treated mTNBC without brain metastases (NCT02574455), SG significantly prolonged OS (median, 12.1 vs 6.7 months) and progression-free survival (PFS; median, 5.6 vs 1.7 months) versus single-agent chemotherapy[8]
 - However, compared with single-agent chemotherapy, SG was associated with increased neutropenia (any grade, 64% vs 44%; grade 3/4, 52% vs 34%), anemia (any grade, 40% vs 28%; grade 3/4, 9% vs 6%), and diarrhea (any grade, 65% vs 17%; grade 3/4, 12% vs 1%)[9]
- The myeloprotective and immune-modulating mechanisms of action of trilaciclib may add meaningful benefit when combined with SG in this patient population
- This study was designed to determine whether administration of trilaciclib prior to SG improves antitumor efficacy and reduces myelotoxicity in patients with mTNBC

METHODS

- This phase 2, single-arm, open-label study was designed to evaluate the safety and efficacy of trilaciclib prior to SG in patients with unresectable, locally advanced TNBC or mTNBC (NCT05113966)
- Key eligibility criteria included:
 - Age ≥ 18 years
 - Confirmed hormone (estrogen and progesterone) receptor–negative and human epidermal growth factor receptor 2–negative status
 - ≥ 2 prior systemic therapies, ≥ 1 of which in the metastatic setting
 - Measurable disease per Response Evaluation Criteria in Solid Tumours version 1.1 (RECIST v1.1)
 - Eastern Cooperative Oncology Group performance status of 0 or 1
 - No known brain metastases at time of enrollment
- Patients received intravenous trilaciclib 240 mg/m^2 immediately prior to SG 10 mg/kg on days 1 and 8 of each 21-day cycle until disease progression or toxicity; prophylactic use of granulocyte colony-stimulating factor was prohibited during cycle 1
- Tumor assessments occurred at screening, every 6 weeks through week 36, then every 9 weeks until disease progression or subsequent anticancer therapy
- **Primary endpoint:** PFS per RECIST v1.1
- **Secondary endpoints**: objective response rate (ORR), clinical benefit rate (CBR; confirmed complete response, partial response, or stable disease lasting ≥ 24 weeks from first dose), duration of response (DOR), OS, myeloprotection, and safety/tolerability

RESULTS

PATIENT DISPOSITION AND CHARACTERISTICS

- As of April 10, 2024, 30 patients had received ≥ 1 dose of any study drug
- Baseline patient demographics and clinical characteristics are summarized in **Table 1**
- One patient remains on study treatment and 12 patients remain in the study
 - The primary reason for treatment discontinuation was disease progression (n = 27)
- Patients received a median (range) of 6.0 (2–29) cycles of treatment, and median follow-up was 15.0 (1–24) months
- Efficacy and study duration results reflect data as of May 7, 2024

TABLE 1. DEMOGRAPHIC AND CLINICAL CHARACTERISTICS AT BASELINE

Characteristic	Patients With mTNBC (N = 30)
Age, years, median (range)	56.0 (30–75)
Female, n (%)	30 (100)
Race, n (%)	
White	26 (86.7)
Black or African American	3 (10.0)
Native Hawaiian or Other Pacific Islander	1 (3.3)
ECOG PS, n (%)	
0	20 (66.7)
1	10 (33.3)
Stage at screening, n (%)	
Locally advanced	2 (6.7)
Metastatic	28 (93.3)
TNBC at initial diagnosis, n (%)	20 (66.7)
Liver metastases, n (%)	11 (36.7)
PD-L1 status,[a] n (%)	
Positive	19 (63.3)
Negative	8 (26.7)
No data	3 (10.0)
***BRCA1/2* mutation status,[a] n (%)**	
Positive	5 (16.7)
Negative	18 (60.0)
No data	7 (23.3)
Prior systemic anticancer regimens, n (%)	
2 or 3	23 (76.7)
> 3	7 (23.3)
Prior anti–PD-(L)1 treatment, n (%)	22 (73.3)
Prior oral CDK4/6i treatment, n (%)	6 (20.0)

[a] PD-L1 and *BRCA* mutation statuses were not required per protocol.
BRCA, breast cancer gene; CDKi, cyclin-dependent kinase inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; mTNBC, metastatic triple-negative breast cancer; PD-1, programmed cell death protein-1; PD-L1, programmed death-ligand 1; TNBC, triple-negative breast cancer.

EFFICACY

- In the overall population (N = 30), confirmed CBR was 46.7% (14/30) and ORR was 23.3% (7/30), with median DOR of 9.1 months
- Median (95% CI) PFS with trilaciclib administered prior to SG was 4.1 (2.2–7.3) months
- Median (95% CI) OS with trilaciclib administered prior to SG was 15.9 (9.9–21.4) months and 12-month OS was 60% (**Table 2**; **Figure 1**)
- Exploratory analyses of OS per key patient subgroups are provided in **Table 2**
 - In the overall population, median (95% CI) OS with censoring at the start of subsequent fam-trastuzumab deruxtecan-nxki (T-DXd) therapy (n = 30) was 17.9 (9.9–not evaluable [NE]) months (**Figure 2**)
 - Among patients with an initial diagnosis of TNBC (n = 20), median OS was 17.9 (9.3–NE) months compared with 12.0 (1.2–NE) months among those without TNBC as the initial diagnosis (n = 10; **Figure 3**)
 - Median OS in patients with prior anti–programmed cell death (ligand) protein-1 (PD-[L]1) treatment (n = 22) was 18.1 (9.3–NE) months compared with 11.4 (1.2–17.9) months in those without prior anti–PD-(L)1 treatment (n = 8; **Figure 4**)
 - In patients without prior oral CDK4/6 inhibitor (CDK4/6i) treatment (n = 24), median OS was 17.9 (9.9–NE) months compared with 8.0 (4.7–NE) months in patients with prior oral CDK4/6i treatment (n = 6; **Figure 5**)

TABLE 2. OS BY PATIENT SUBGROUP

Patient Subgroup	Number of Events/ Number of Patients	Median (95% CI) OS, Months
Overall	19/30	15.9 (9.9–21.4)
Age, years		
< 65	13/23	17.9 (9.3–NE)
≥ 65	6/7	10.8 (3.8–18.1)
Race		
White	16/26	16.3 (9.9–NE)
Black	2/3	8.5 (5.4–NE)
Native Hawaiian or Other Pacific Islander	1/1	14.2 (NE–NE)
TNBC at initial diagnosis		
Yes	12/20	17.9 (9.3–NE)
No	7/10	12.0 (1.2–NE)
Liver metastasis at baseline		
Yes	8/11	9.9 (5.4–NE)
No	11/19	18.1 (9.9–NE)
Prior systemic anticancer regimens		
2 or 3	15/23	16.3 (9.9–21.4)
> 3	4/7	10.8 (4.7–NE)
Prior anti–PD-(L)1 treatment		
Yes	12/22	18.1 (9.3–NE)
No	7/8	11.4 (1.2–17.9)
Prior oral CDK4/6i treatment[a]		
Yes	5/6	8.0 (4.7–NE)
No	14/24	17.9 (9.9–NE)
SACT with T-DXd		
Yes	6/9	14.2 (4.7–NE)
No	13/21	17.1 (9.3–NE)

[a] Patients with an initial diagnosis of hormone receptor–positive breast cancer may have received prior oral CDK4/6i treatment.
CDKi, cyclin-dependent kinase inhibitor; NE, not evaluable; OS, overall survival; PD-1, programmed cell death protein-1; PD-L1, programmed death-ligand 1; SACT, subsequent anticancer therapy; T-DXd, fam-trastuzumab deruxtecan-nxki; TNBC, triple-negative breast cancer.

SAFETY AND TOLERABILITY

- Safety data are summarized in **Table 3**
- Any-grade and grade 3/4 treatment-emergent adverse events (TEAEs) were reported in 96.7% and 56.7% of patients, respectively
 - The most common any-grade TEAEs, occurring in > 20% of patients, were fatigue (63.3%), nausea (53.3%), and diarrhea (43.3%); overall, concomitant medicine was used for diarrhea management in 20% of patients
- Hematologic adverse events (AEs) of any grade and grade 3/4 were reported in 53.3% and 30.0% of patients, respectively
 - Any-grade and grade 3/4 neutropenia were reported in 40.0% and 23.3% of patients, respectively; overall, 23.3% of patients received granulocyte colony-stimulating factor
- Serious AEs, regardless of causality, were reported in 6/30 (20%) patients; none were fatal and none of the event terms occurred in > 1 patient

FIGURE 1. KAPLAN-MEIER CURVE OF OS IN THE OVERALL POPULATION (N = 30)

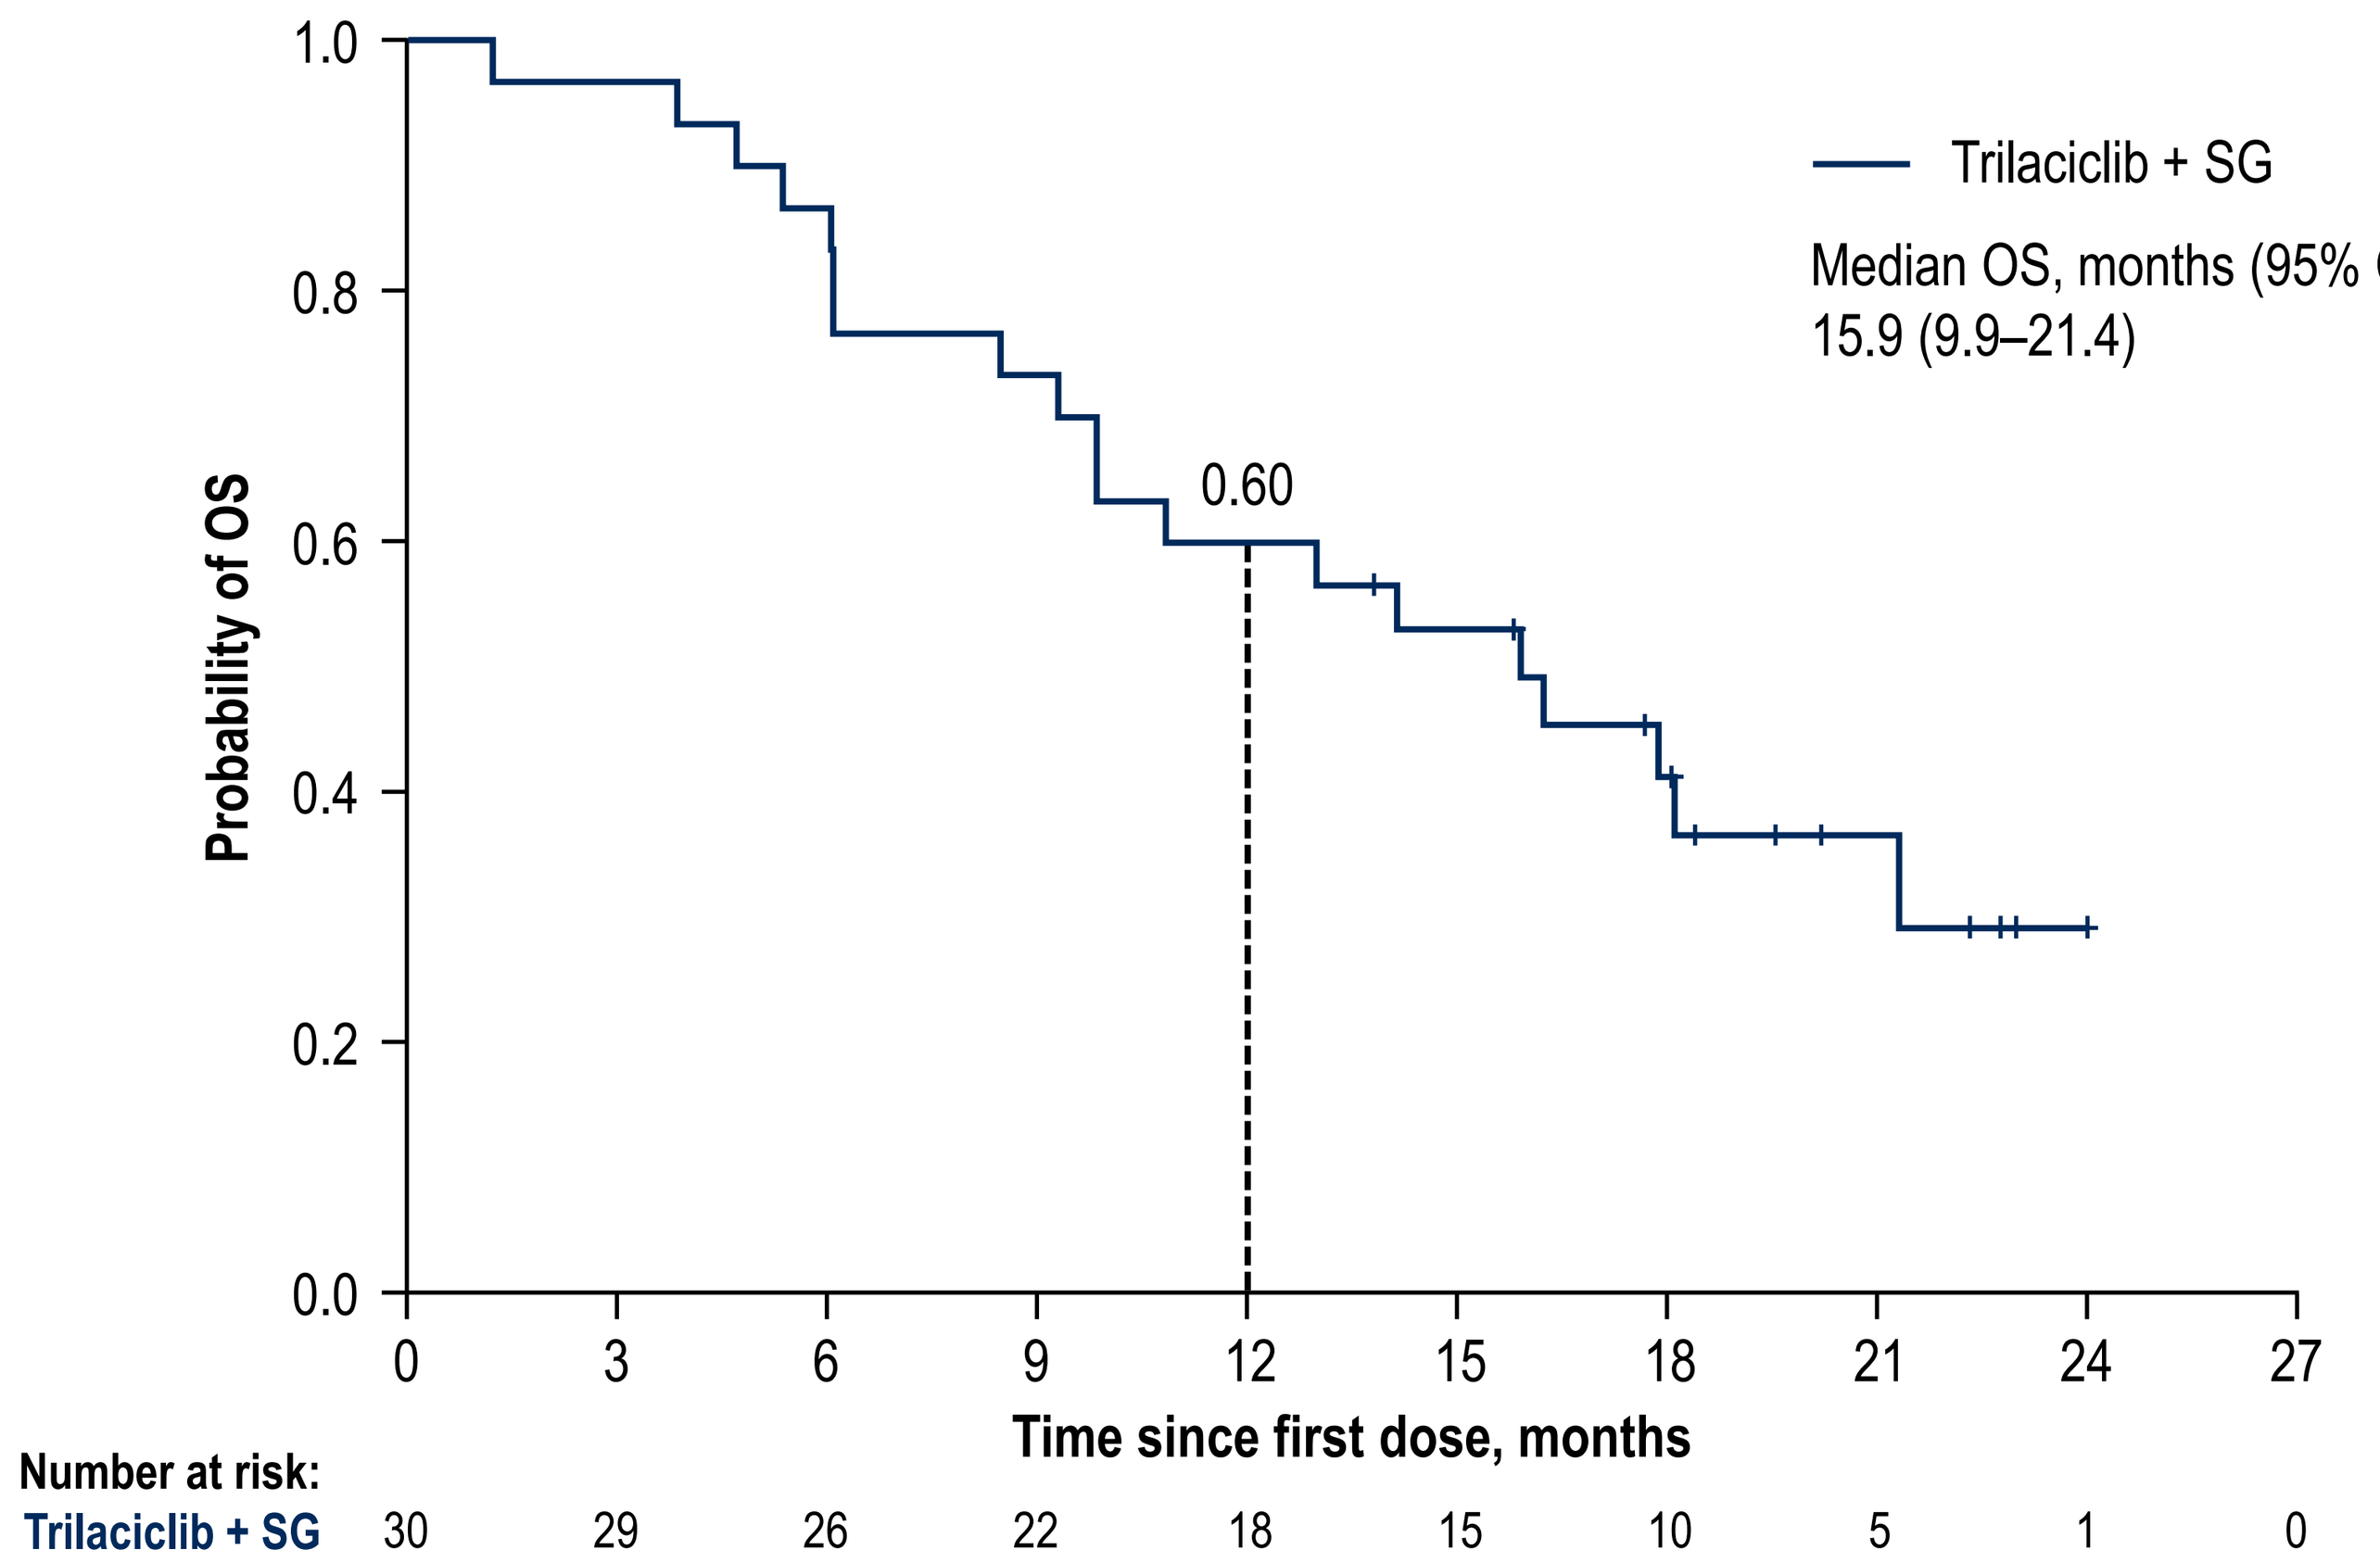
Trilaciclib + SG
Median OS, months (95% CI):
15.9 (9.9–21.4)
0.60
Probability of OS
1.0 0.8 0.6 0.4 0.2 0.0
0 3 6 9 12 15 18 21 24 27
Time since first dose, months
Number at risk:
Trilaciclib + SG 30 29 26 22 18 15 10 5 1 0

The dashed line shows 12-month OS.
OS, overall survival; SG, sacituzumab govitecan.

FIGURE 2. KAPLAN-MEIER CURVE OF OS WITH CENSORING AT THE START OF SUBSEQUENT T-DXD THERAPY (N = 30)

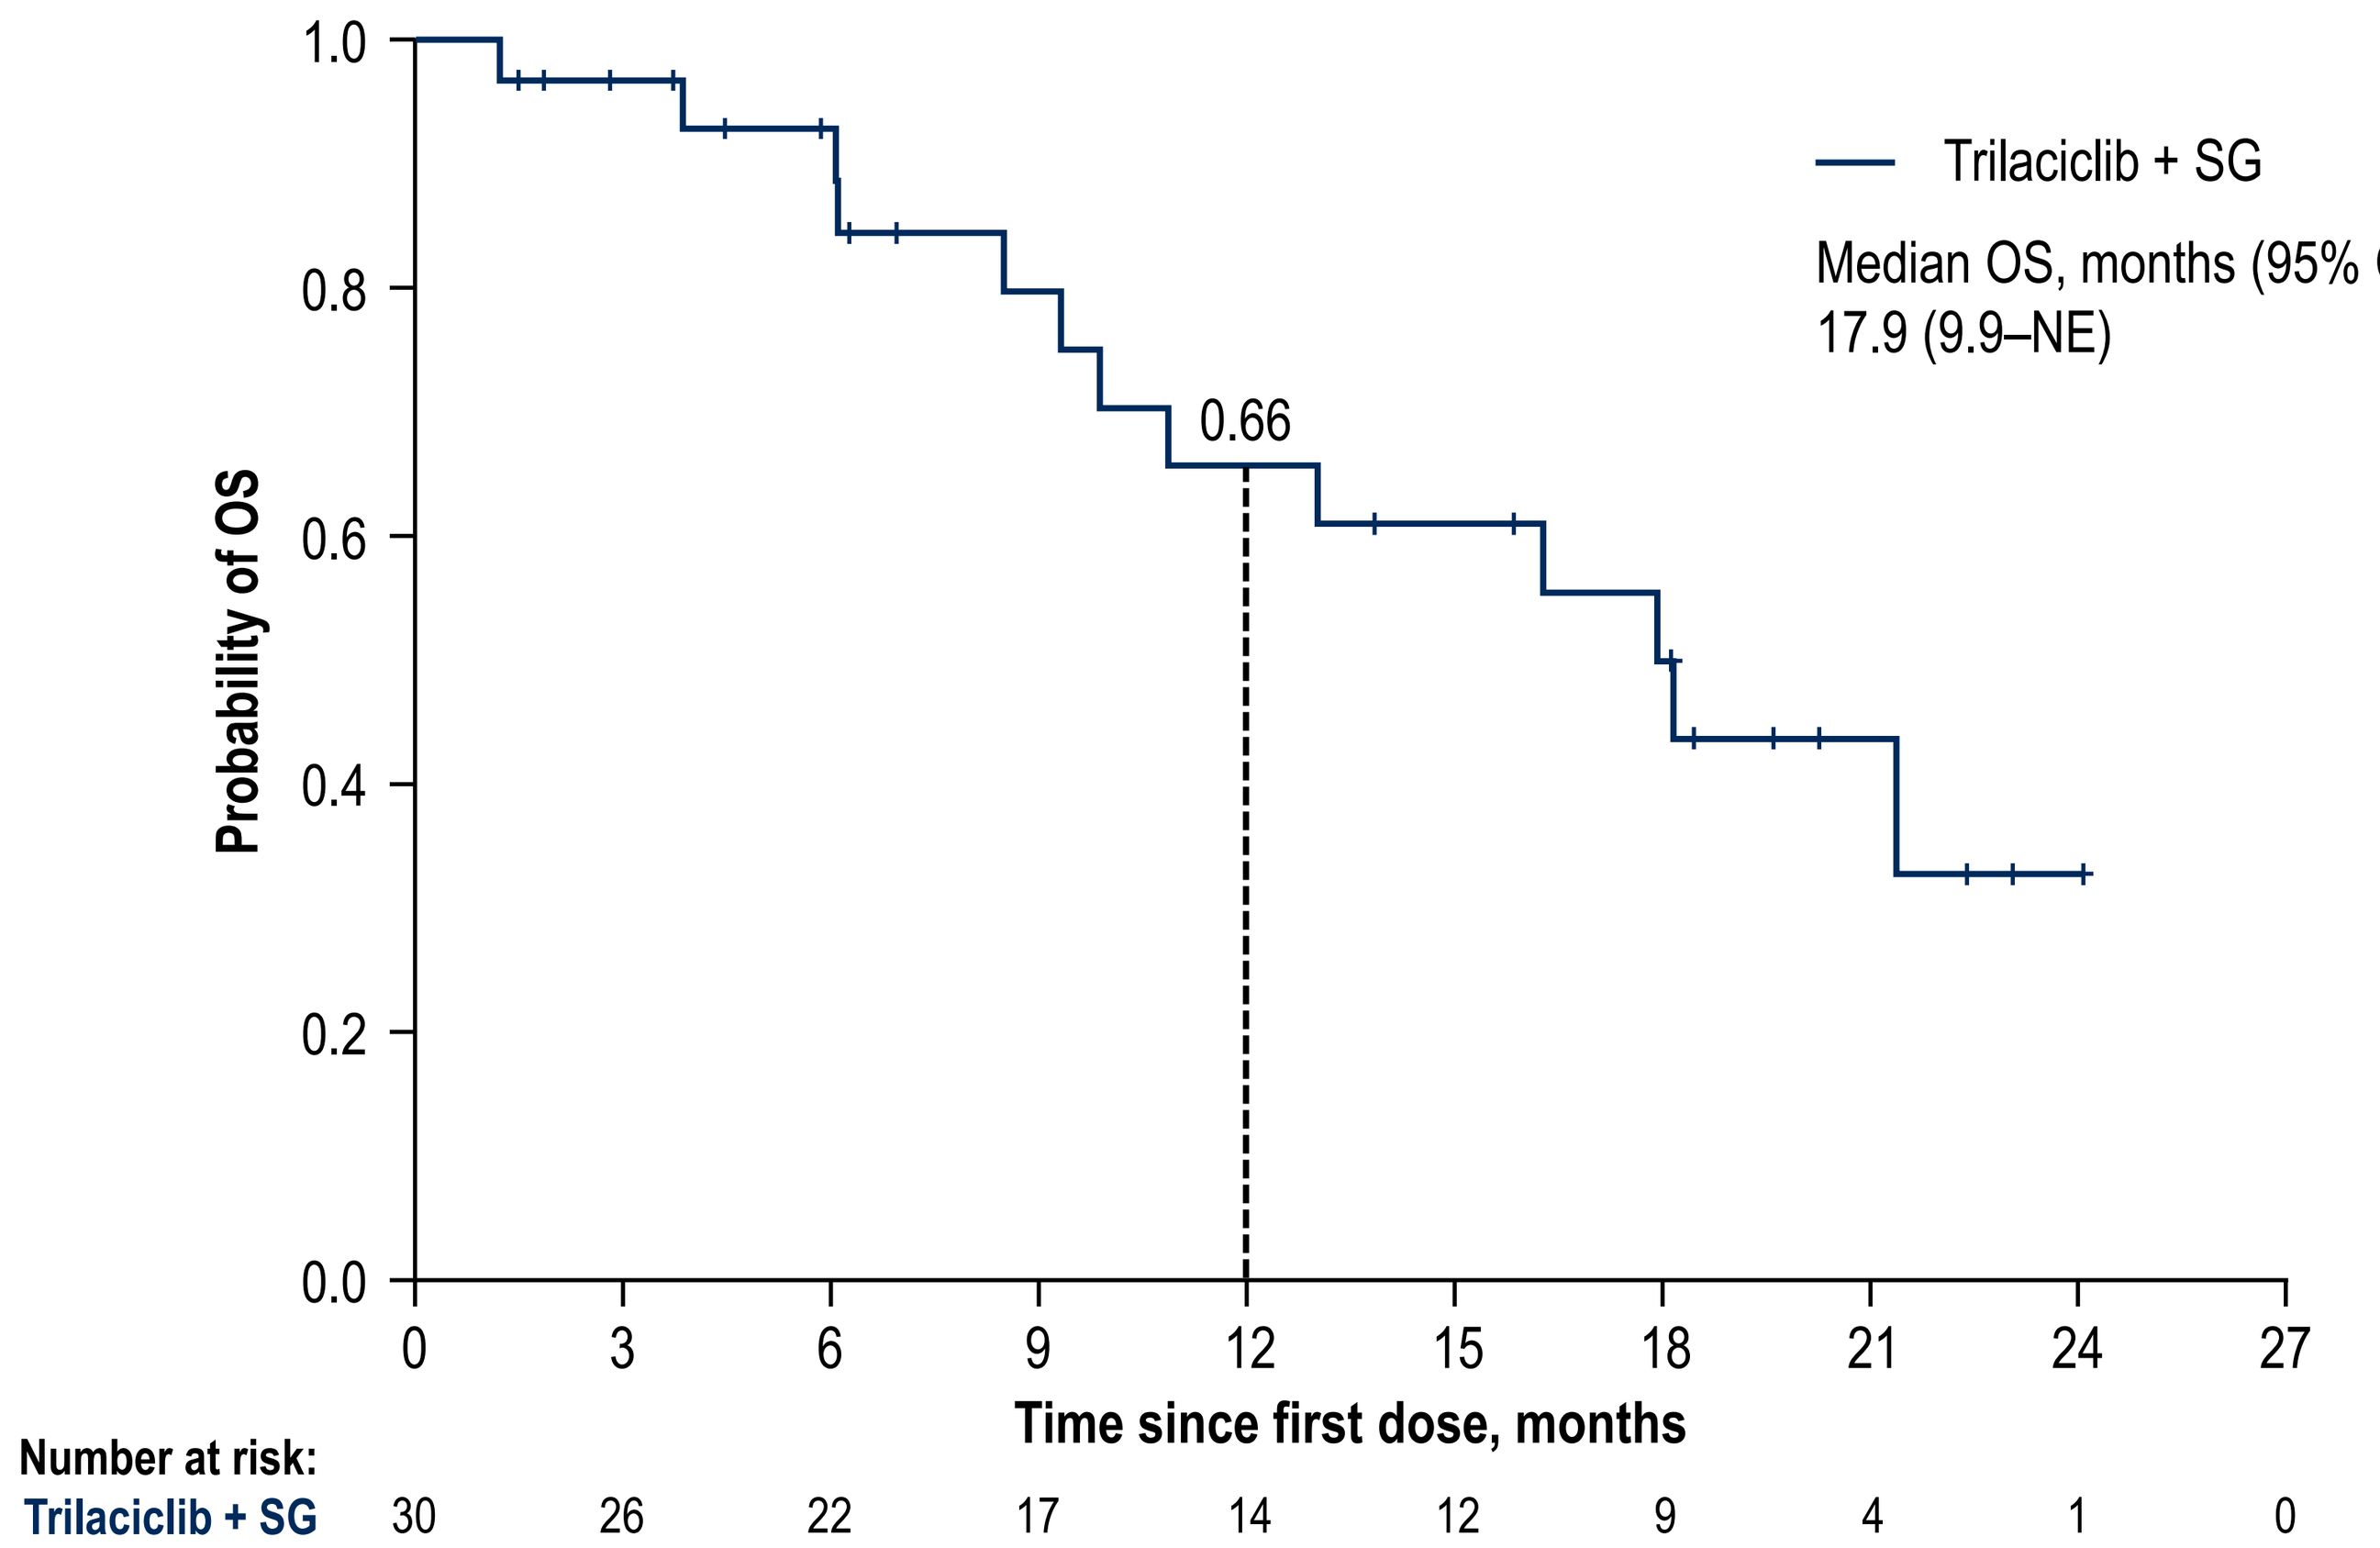
Trilaciclib + SG
Median OS, months (95% CI):
17.9 (9.9–NE)
0.66
Probability of OS
1.0 0.8 0.6 0.4 0.2 0.0
0 3 6 9 12 15 18 21 24 27
Time since first dose, months
Number at risk:
Trilaciclib + SG 30 26 22 17 14 12 9 4 1 0

The dashed line shows 12-month OS.
NE, not evaluable; OS, overall survival; SG, sacituzumab govitecan; T-DXd, fam-trastuzumab deruxtecan-nxki.

FIGURE 3. KAPLAN-MEIER CURVE OF OS BY INITIAL DIAGNOSIS OF TNBC

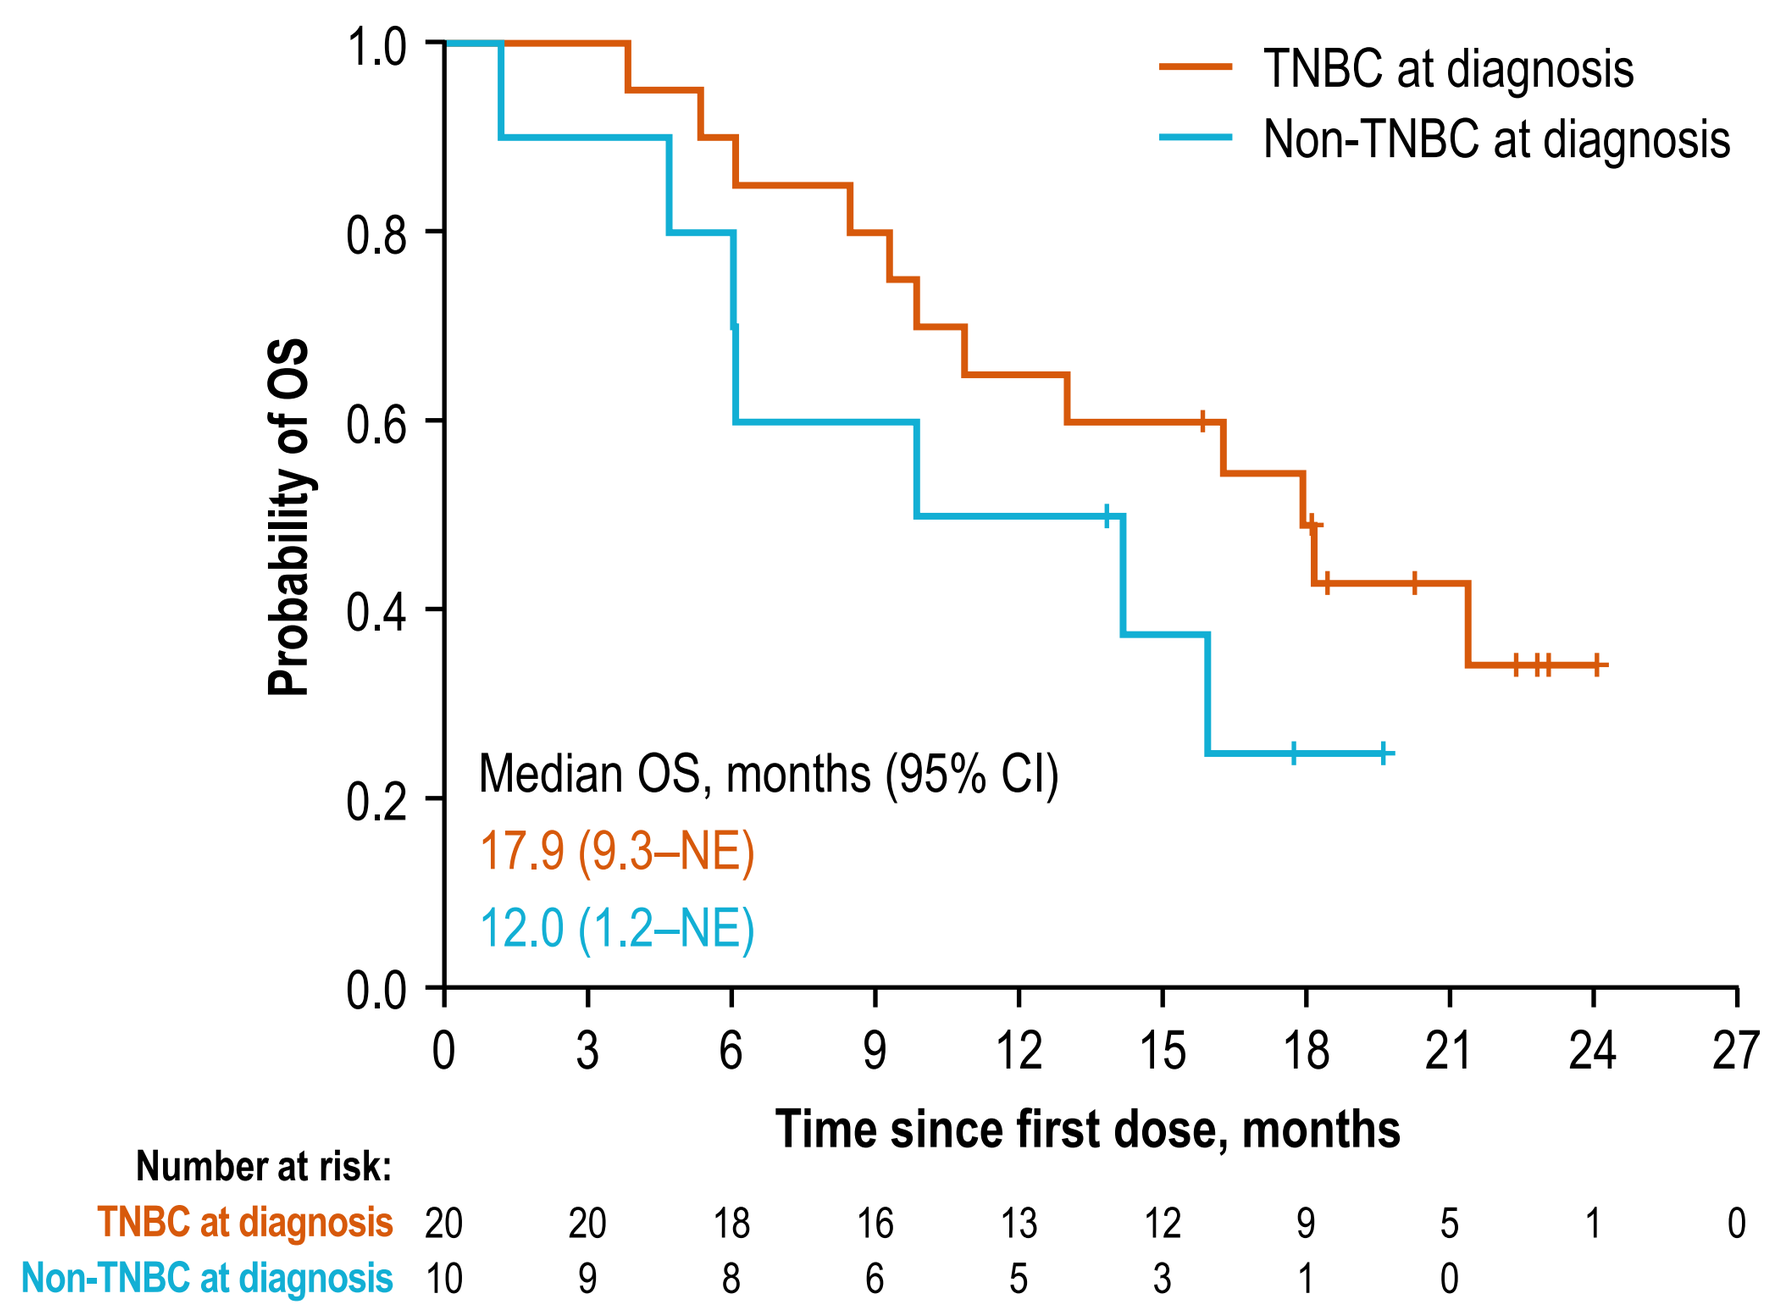
TNBC at diagnosis
Non-TNBC at diagnosis
Median OS, months (95% CI)
17.9 (9.3–NE)
12.0 (1.2–NE)
Probability of OS
1.0 0.8 0.6 0.4 0.2 0.0
0 3 6 9 12 15 18 21 24 27
Time since first dose, months
Number at risk:
TNBC at diagnosis 20 20 18 16 13 12 9 5 1 0
Non-TNBC at diagnosis 10 9 8 6 5 3 1 0

NE, not evaluable; OS, overall survival; TNBC, triple-negative breast cancer.

FIGURE 4. KAPLAN-MEIER CURVE OF OS BY PRIOR ANTI–PD-(L)1 TREATMENT

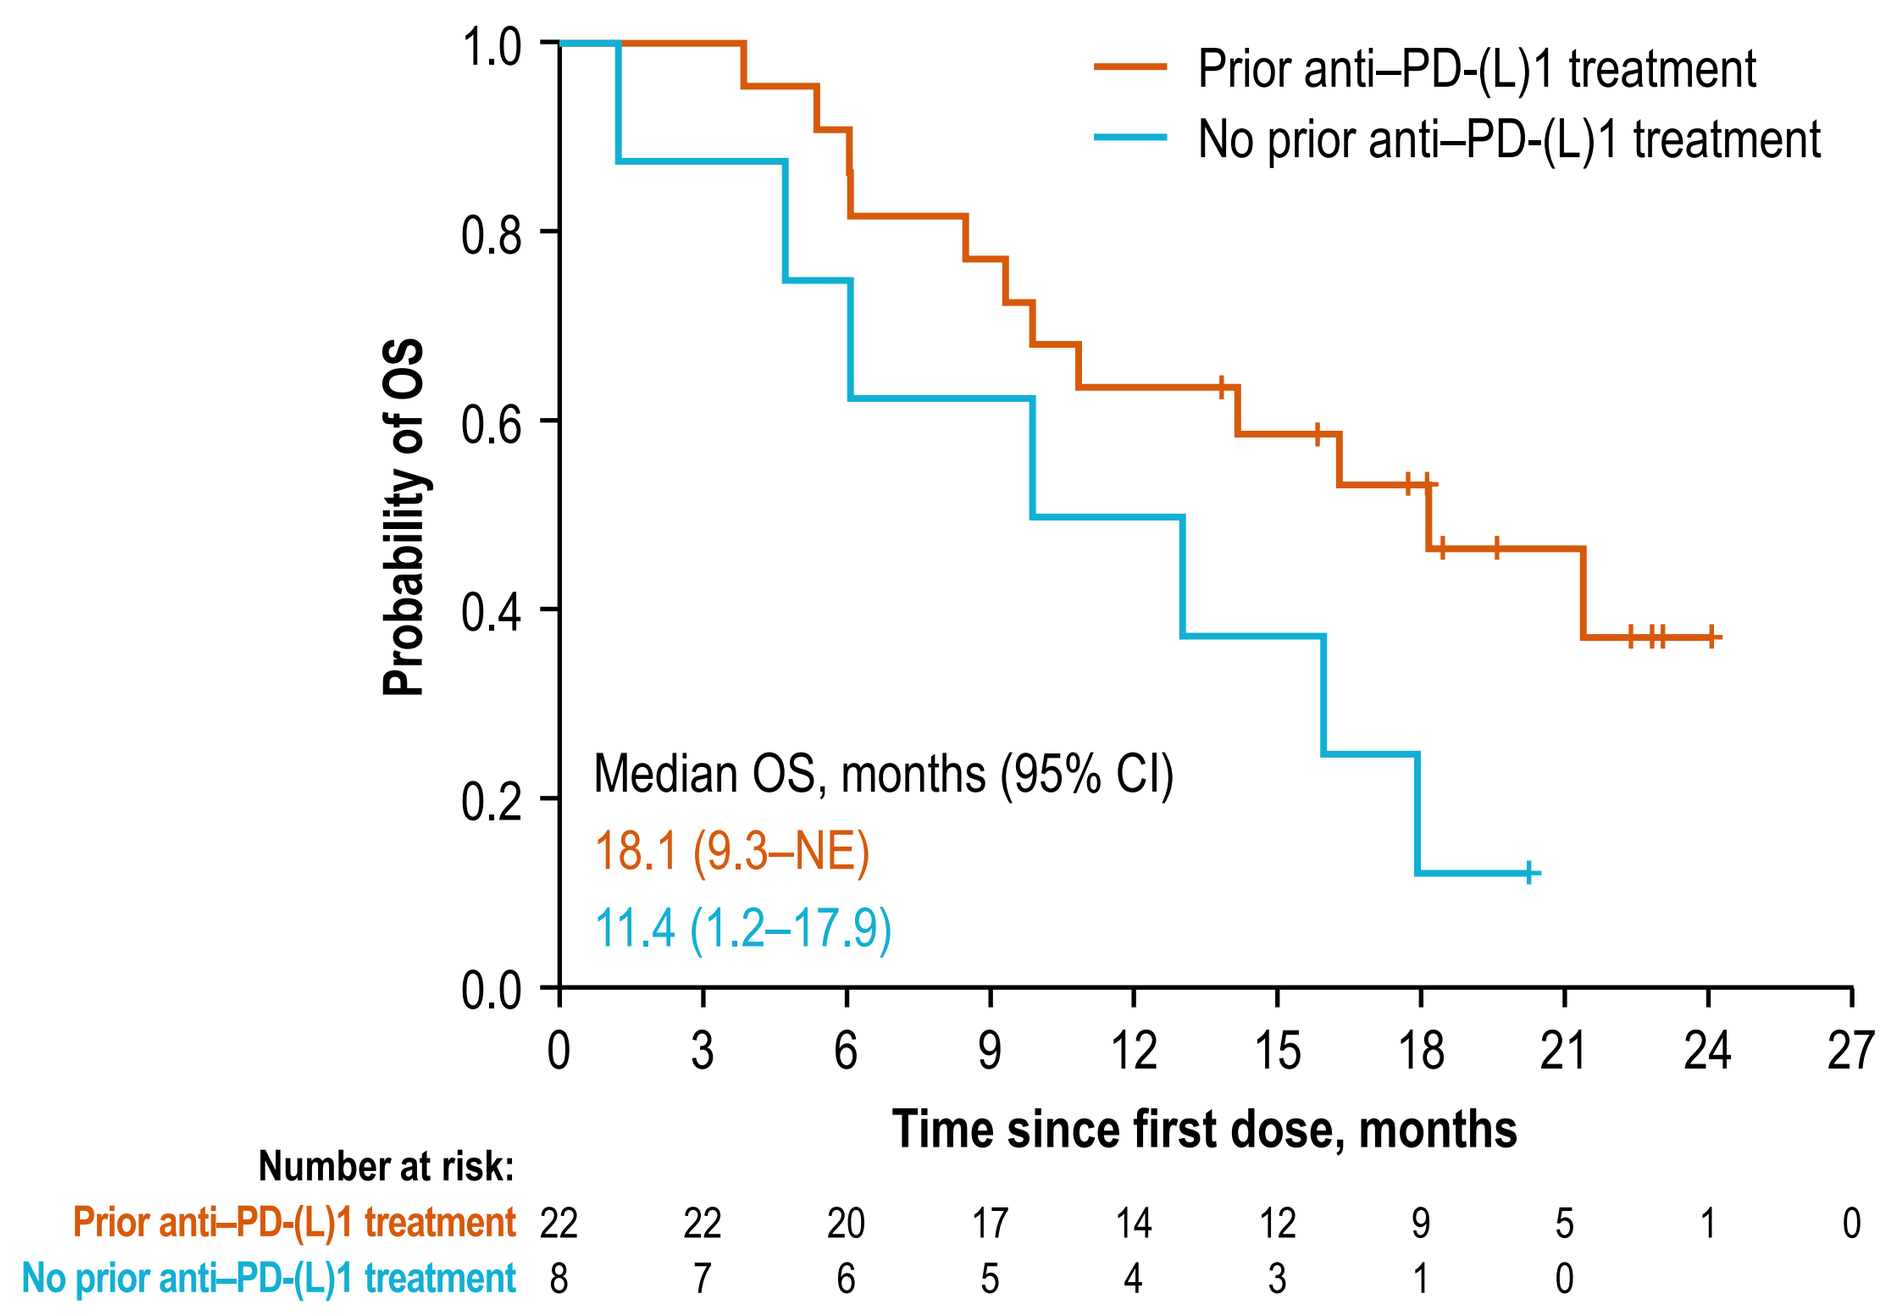
Prior anti–PD-(L)1 treatment
No prior anti–PD-(L)1 treatment
Median OS, months (95% CI)
18.1 (9.3–NE)
11.4 (1.2–17.9)
Probability of OS
1.0 0.8 0.6 0.4 0.2 0.0
0 3 6 9 12 15 18 21 24 27
Time since first dose, months
Number at risk:
Prior anti–PD-(L)1 treatment 22 22 20 17 14 12 9 5 1 0
No prior anti–PD-(L)1 treatment 8 7 6 5 4 3 1 0

NE, not evaluable; OS, overall survival; PD-1, programmed cell death protein-1; PD-L1, programmed death-ligand 1.

FIGURE 5. KAPLAN-MEIER CURVE OF OS BY PRIOR ORAL CDK4/6i TREATMENT

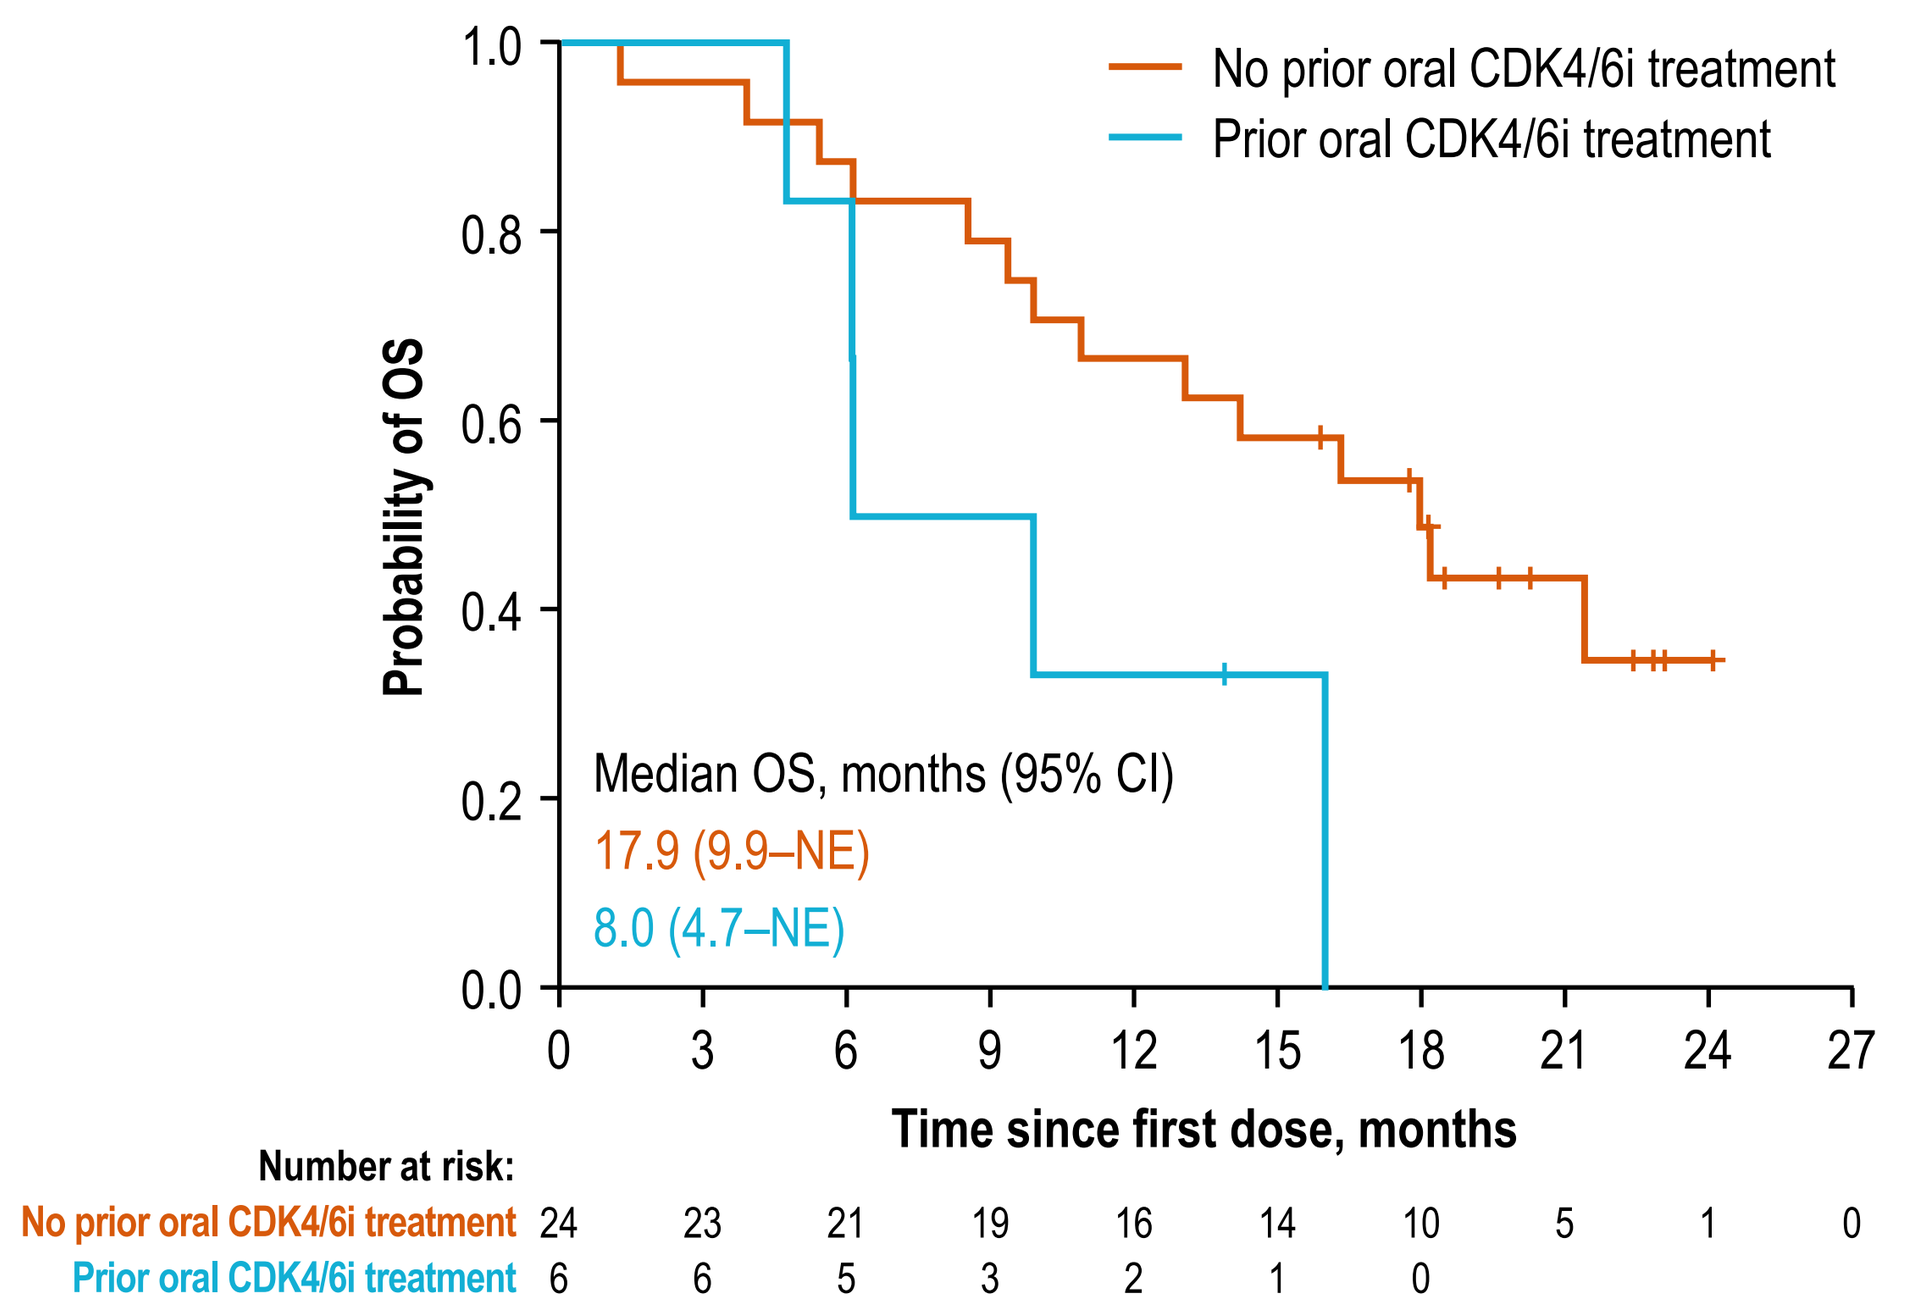
No prior oral CDK4/6i treatment
Prior oral CDK4/6i treatment
Median OS, months (95% CI)
17.9 (9.9–NE)
8.0 (4.7–NE)
Probability of OS
1.0 0.8 0.6 0.4 0.2 0.0
0 3 6 9 12 15 18 21 24 27
Time since first dose, months
Number at risk:
No prior oral CDK4/6i treatment 24 23 21 19 16 14 10 5 1 0
Prior oral CDK4/6i treatment 6 6 5 3 2 1 0

CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; NE, not evaluable; OS, overall survival.

TABLE 3. SUMMARY OF TEAES AND HEMATOLOGIC TEAES

	Any Grade (N = 30)	Grade 3/4 (N = 30)
Patients with ≥ 1 TEAE,[a] n (%)	29 (96.7)	17 (56.7)
Fatigue	19 (63.3)	1 (3.3)
Nausea	16 (53.3)	1 (3.3)
Diarrhea	13 (43.3)	2 (6.7)
Alopecia	11 (36.7)	0
Headache	10 (33.3)	0
Constipation	9 (30.0)	0
Vomiting	8 (26.7)	1 (3.3)
Dizziness	7 (23.3)	0
Insomnia	7 (23.3)	0
Patients with ≥ 1 hematologic TEAE, n (%)	16 (53.3)	9 (30.0)
Neutropenia[b]	12 (40.0)	7 (23.3)
Leukopenia[c]	10 (33.3)	4 (13.3)
Anemia[d]	3 (10.0)	1 (3.3)
Thrombocytopenia[e]	0	0

[a] TEAE occurring in > 20% of patients.
[b] Category includes neutropenia and decreased neutrophil count.
[c] Category includes leukopenia and decreased white blood cell count.
[d] Category includes anemia, macrocytic anemia, decreased red blood cell count, and decreased hemoglobin.
[e] Category includes thrombocytopenia and decreased platelet count.
TEAE, treatment-emergent adverse event.

CONCLUSIONS

- A median OS of 15.9 months compares favorably with historical data for SG alone in previously treated patients with mTNBC[8,9]
- In an exploratory analysis, median OS in the overall population with censoring at the start of subsequent T-DXd therapy was longer than without censoring
 - The outcomes in this censored population are potentially more comparable with results from the ASCENT study, as T-DXd was not approved for human epidermal growth factor receptor 2–low breast cancer at the time of ASCENT
- Exploratory analyses of OS in other patient subgroups showed that prolonged OS was observed among patients with an initial diagnosis of TNBC versus non-TNBC, prior anti–PD-(L)1 treatment versus no prior anti–PD-(L)1, and no prior oral CDK4/6i treatment versus prior oral CDK4/6i
- Although cross-trial comparisons should be made with caution, administering trilaciclib prior to SG reduced the frequency of multiple AEs, notably neutropenia, anemia, and diarrhea, compared with results from a historical trial of SG alone[9]
- Results from this study support further evaluation of trilaciclib administered prior to SG or other antibody–drug conjugates

American Society of Clinical Oncology (ASCO) Annual Meeting | May 31 – June 4, 2024 | Chicago, IL, USA

REFERENCES:
1. COSELA® (trilaciclib). Prescribing Information. https://www.g1therapeutics.com/cosela/pi/. Accessed April 18, 2024.
2. Daniel D, et al. *Int J Cancer*. 2021;148:2557–70.
3. Weiss JM, et al; G1T28-02 Study Group. *Ann Oncol*. 2019;30:1613–21.
4. Hart LL, et al. *Adv Ther*. 2021;38:350–65.
5. Tan AR, et al. *Breast Cancer Res Treat*. 2023;201:307–16.
6. Tan AR, et al. *Clin Cancer Res*. 2022;28:629–36.
7. TRODELVY® (sacituzumab govitecan-hziy). Prescribing Information. https://www.gilead.com/-/media/files/pdfs/medicines/oncology/trodelvy/trodelvy_pi.pdf. Accessed April 18, 2024.
8. Bardia A, et al; ASCENT Clinical Trial Investigators. *N Engl J Med*. 2021;384:1529–41.
9. Bardia A, et al. *J Clin Oncol*. 2024: Online ahead of print. doi: 10.1200/JCO.23.01409.

ACKNOWLEDGMENTS:
- We thank all the investigators and site staff, with special thanks to the patients and their families.
- We thank Jingshan Zhang for his support in the preparation of the abstract.
- Study sponsored by G1 Therapeutics, Inc. Medical writing assistance was provided by Farhana Burnett, PhD, CMPP, of Envision Pharma Group, funded by G1 Therapeutics, Inc.

CONTACT INFORMATION:
If you have any questions or comments, please contact Andrew Beelen at abeelen@g1therapeutics.com